Poster presented at the 9th Joint ECTRIMS-ACTRIMS Meeting, 11–13 October 2023 Milan, Italy.

Incidence of COVID-19 in patients with multiple sclerosis who received SARS-CoV-2 vaccines and are under treatment with high-efficacy therapies in Argentina

P1573

Edgar Carnero Contentti*[1], Pablo A. López[1], Agustín Pappolla[2], Ricardo Alonso[3,4], Berenice Silva[2,3], Norma Deri[5,6], María E. Balbuena[7], Marcos Burgos[8], Geraldine Luetic[9], Amelia Alvez Pinheiro[10], Mariela Cabrera[11], Javier Hryb[12], Pedro Nofal[13], Claudia Pestchanker[14], Carlos Vrech[15,] Darío Tavolini[16], Verónica Tkachuk[7], Gisela Zanga[17], Mariano Marrodan[18], María Célica Ysrraelit[18], Jorge Correale[18], Adriana Carrá[19,20], Belén Federico[21], Orlando Garcea[3], Nora Fernandez Liguori[4], Liliana Patrucco[22], Edgardo Cristiano[22], Diego Giunta[23], Marina Alonso Serena[23], Juan I. Rojas[22,24]* on behalf RelevarEM

1.Department of Neurosciences, Neuroimmunology Unit, Hospital Aleman, Buenos Aires, Argentina, 2.Servicio de Neurología, Hospital Italiano de Buenos Aires, Buenos Aires, Argentina; 3.Hospital Ramos Mejía, Centro Universitario de Esclerosis Múltiple, CABA, Argentina; 4.Sanatorio Güemes, CABA, Argentina; 5. Centro de Investigaciones Diabaid, CABA, Argentina; 6.Hospital Fernandez, Buenos Aires, Argentina; 7.Sección de Neuroinmunología y Enfermedades Desmielinizantes, Servicio de Neurología, Hospital de Clínicas José de San Martín, CABA, Argentina; 8.Servicio de Neurología, Hospital San Bernardo, Salta, Argentina; 9.Instituto de Neurociencias de Rosario, San Lorenzo, Rosario, Santa Fé, Argentina; 10.Hospital San Martín, Paraná, Entre Ríos, Argentina; 11. Hospital Militar Campo de Mayo, Buenos Aires, Argentina; 12.Servicio de Neurología, Hospital Carlos G Durand, Buenos Aires, Argentina; 13.Hospital de Clínicas Nuestra Señora ˜ del Carmen, Tucumán, Argentina; 14. Neurology Department, Hospital Central Dr. Ramon Carrillo, San Luis; 15.Departamento de Enfermedades desmielinizantes, Sanatorio Allende, Córdoba, Argentina; 16. INECO Neurociencias Oroño, Rosario, Santa Fe, Argentina; 17.Departamento de Neurología, Hospital Cesar Milstein, Buenos Aires, Argentina; 18.Departamento de Neurología, FLENI, CABA, Argentina; 19.Sección de Enfermedades Desmielinizantes, Hospital Británico, CABA, Argentina; 20.Hospital Universitario Fundación Favaloro, Buenos Aires, Argentina; 21.Novartis SRL, Argentina; 22.Centro de esclerosis múltiple de Buenos Aires, Buenos Aires, Argentina; 23.Servicio de clínica médica, Hospital Italiano de Buenos Aires, CABA, Argentina; 24.Servicio de Neurología, Unidad de EM y Enfermedades Desmielinizantes, Hospital Universitario de CEMIC, Buenos Aires, Argentina

SUMMARY

- The effectiveness of SARS-CoV-2 vaccines in PwMS is focused on mRNA vaccines (the type of vaccines not widely implemented in Argentina),
- It is difficult to extrapolate results from international research studies into Argentina's context
- We found an incidence density of breakthrough SARS_CoV-2 infection of 3.5 per 10,000 patients/day (95%CI 2.3-6.7) after vaccination in Argentina mainly with Sputnik or Astra Zeneca vaccines.
- We described the incidence rate after vaccination for every HET used, it being significantly higher for ocrelizumab compared with other HETs.
- We also observed a decrease in the frequency of COVID-19 infection between pre- and post-vaccination in the included HET patients.

INTRODUCTION

- Different vaccines are being used to achieve immunization against severe acute respiratory syndrome coronavirus 2 (SARS-CoV-2) worldwide.
- As data generated regarding the effectiveness of SARS-CoV-2 vaccines in PwMS is focused on mRNA vaccines (the type of vaccines not widely implemented in Argentina), it is difficult to extrapolate results from international research studies into Argentina context and the need of local data is urgent and necessary.

OBJECTIVE

- We aimed to evaluate the incidence of SARS-CoV-2 breakthrough infection and the side effects of SARS-CoV-2 vaccines in people with MS (PwMS) on high-efficacy disease-modifying therapies (DMTs) (HET) included in the national MS registry in Argentina (RelevarEM).

METHODS

- Non-interventional, retrospective cohort study that collected information directly from RelevarEM (clinical trials registry number NCTNCT03375177). Adult PwMS who had been treated for at least 6 months with a HET (ocrelizumab, natalizumab, alemtuzumab, cladribine) who had received at least 2 doses of SARS-CoV-2 vaccines available in Argentina were included. A full course of vaccination was considered after the second dose of the corresponding vaccines. A positive COVID-19 case was defined according to the definition established by the Ministry of Health in Argentina.
- The primary outcome was to assess the incidence of SARS-CoV-2 breakthrough infection in PwMS on HET vaccinated for COVID-19. Clinical activity after the first dose and up to 6 months after full course vaccine was also collected. Cumulative incidence of SARS-CoV-2 infection was reported for the whole cohort by Kaplan Meier survival curves (which is expressed in percentage) as well as incidence density (which is expressed per 10.000 patient/days with 95%CI).

RESULTS

- 228 PwMS were included. Most frequent first and second dose received was Astra-Zeneca vaccine, followed by Sputnik vaccine. Most frequent HETs used in included patients were cladribine in 79 (34.8%), as shown in **Table 1**.
- A total of 15 positive COVID-19 cases were observed for a total exposure time of 43,180 patient/days and 4 cases of hospitalization due to severe SARS-CoV-2 infection). We found an incidence density of breakthrough COVID-19 infection of 3.5 x 10.000 patients/day (95%CI 2.3-6.7) after vaccination in Argentina. We described the incidence rate after vaccination for every HET used, it being significantly higher for ocrelizumab compared with other HETs (p=0.005). Only 5 patients presented a relapse during the follow-up period with no differences regarding the pre-vaccination period.

Figure 1. Incidence density of COVID-19 infections post- and pre-vaccination

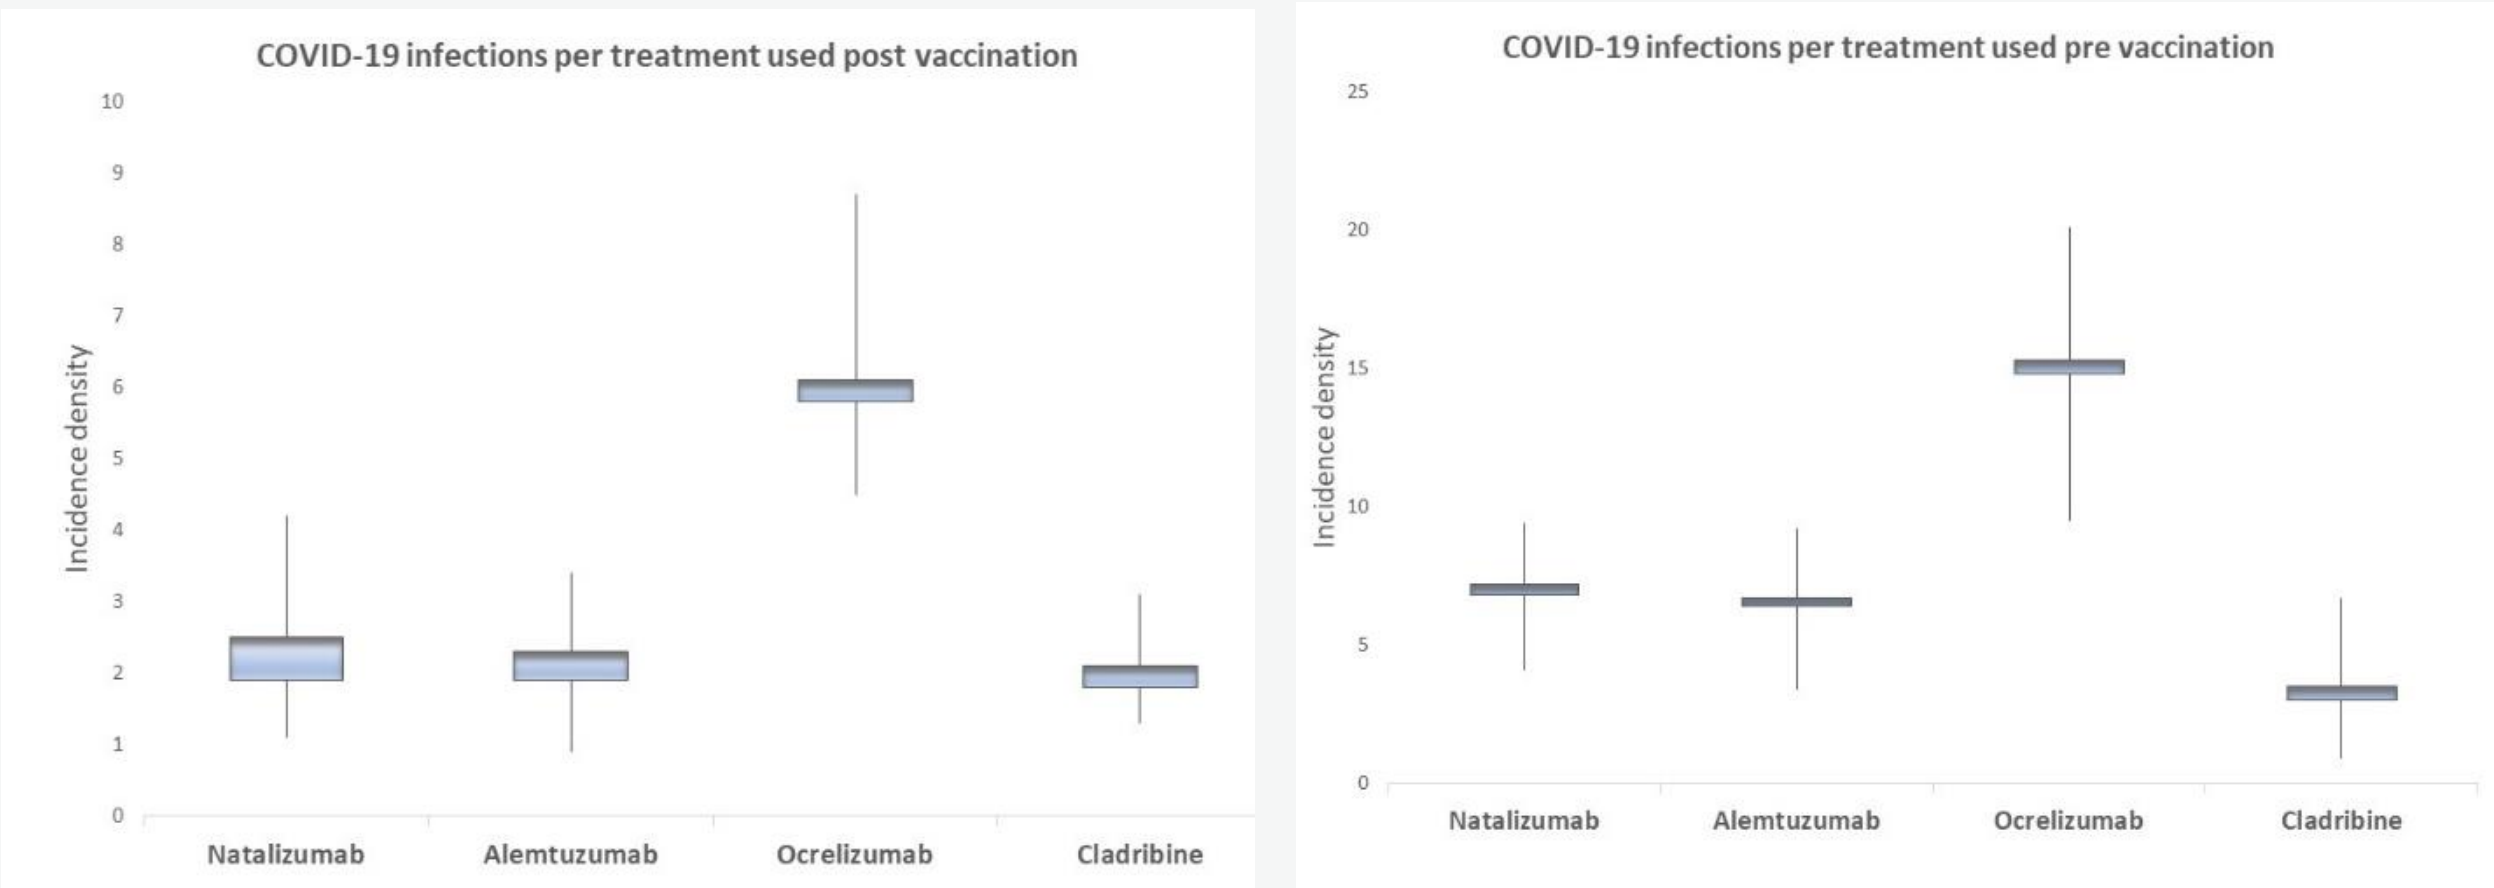
COVID-19 infections per treatment used post vaccination
Incidence density
10
9
8
7
6
5
4
3
2
1
0
Natalizumab
Alemtuzumab
Ocrelizumab
Cladribine
COVID-19 infections per treatment used pre vaccination
Incidence density
25
20
15
10
5
0
Natalizumab
Alemtuzumab
Ocrelizumab
Cladribine

Figure 2. Pre- and post-vaccination comparison of incidence density infection with COVID 19 in included patients.

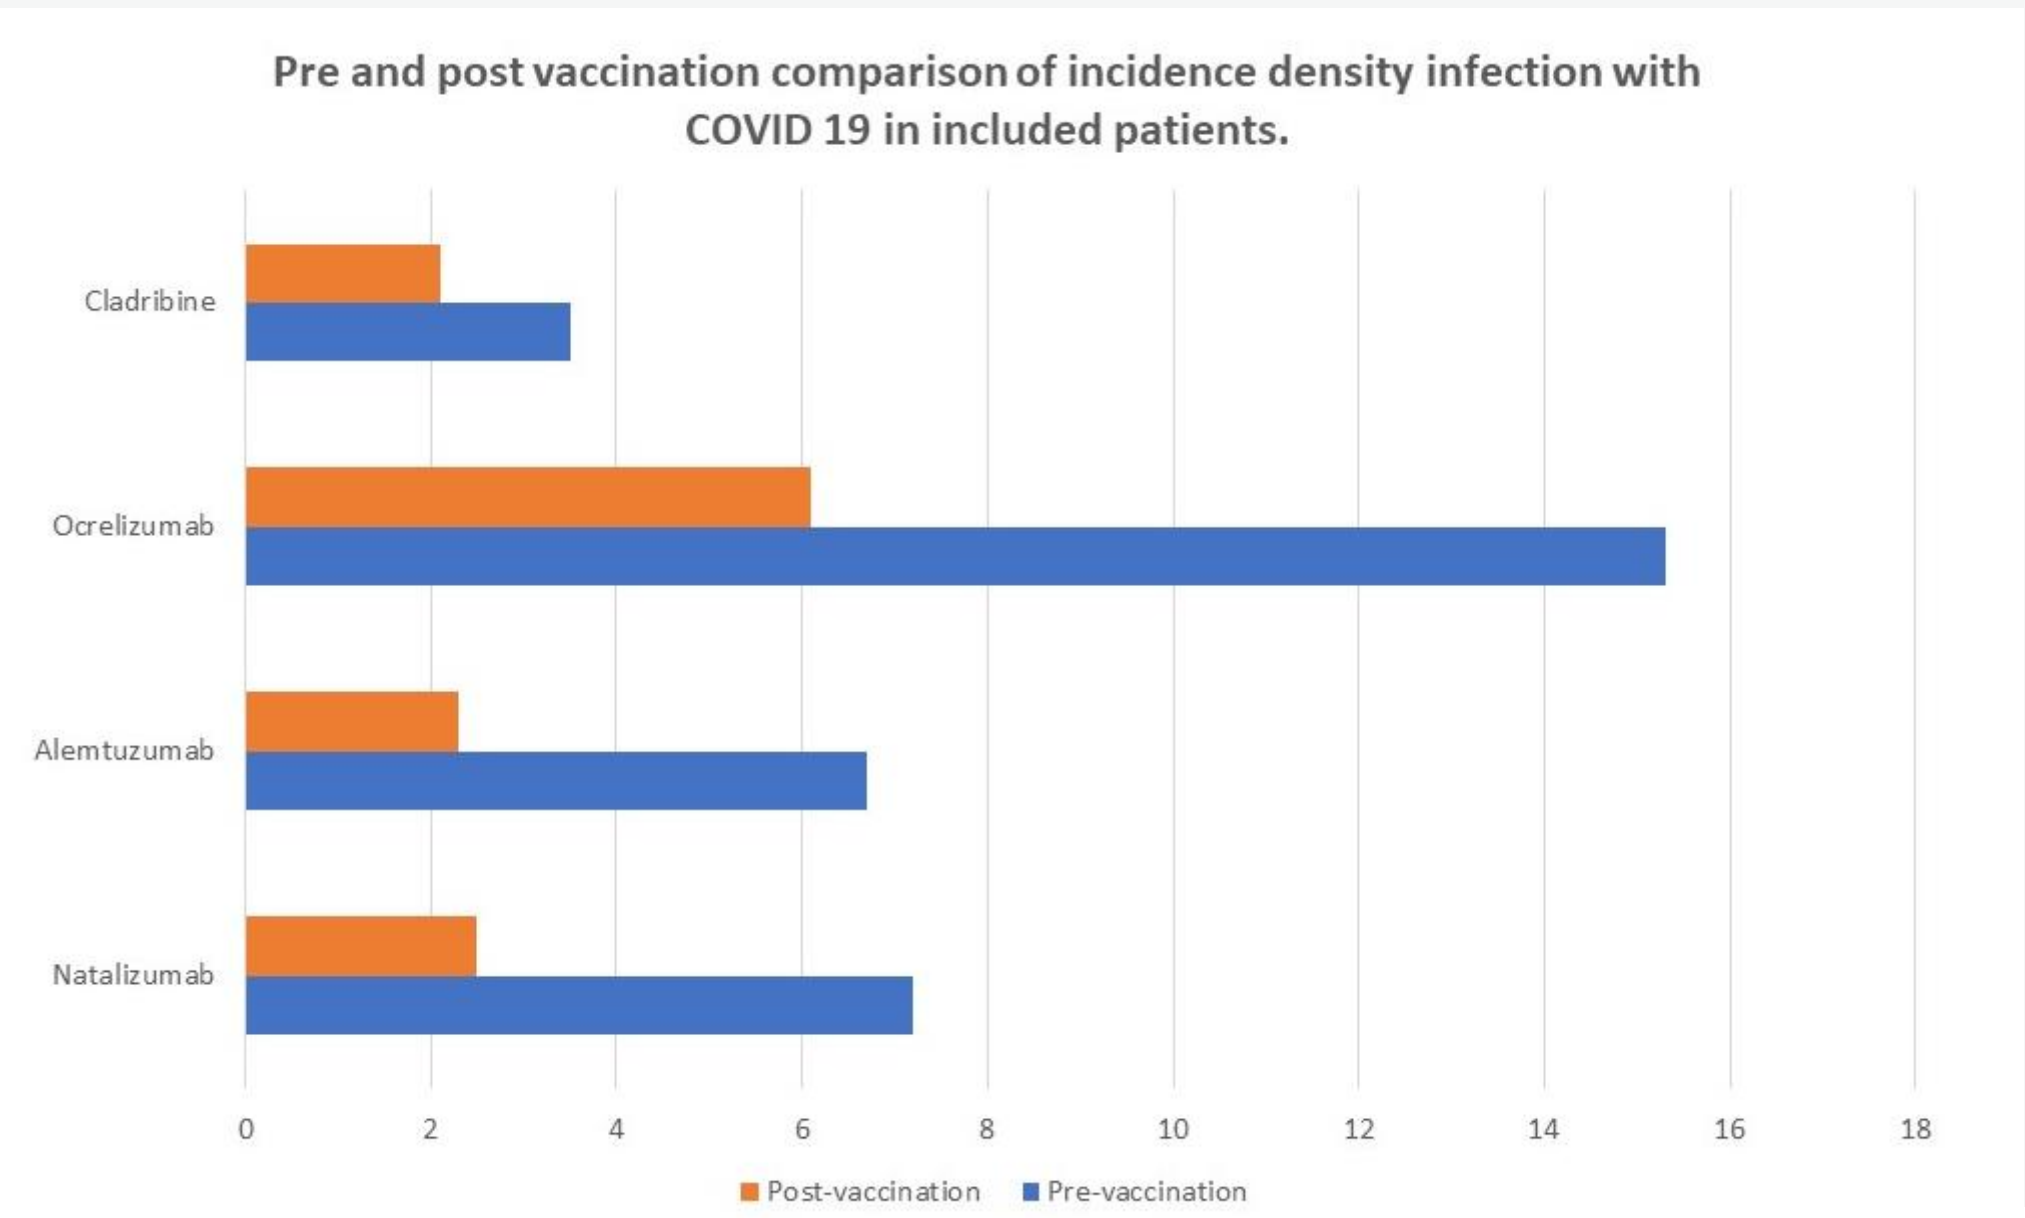
Pre and post vaccination comparison of incidence density infection with COVID 19 in included patients.
Cladribine
Ocrelizumab
Alemtuzumab
Natalizumab
0
2
4
6
8
10
12
14
16
18
Post-vaccination
Pre-vaccination

Table 1. Baseline characteristics, follow-up time, and vaccines received

	N = 228
Mean age at study entry, years (SD)	38.7± 8.5
Mean disease duration, years (SD)	7.6± 5.5
RRMS, n (%)	196 (86)
SPMS, n (%)	32 (14)
Female gender, n (%)	166 (72.8)
Median EDSS at Study entry, (IQR)(Range)	3 (2.0) (1-7)
ARR in the 6 months previous first dose of vaccine	0.32 ± 0.18
Disease modifying treatment (DMT)* Natalizumab, n (%) Alemtuzumab, n (%) Ocrelizumab, n (%) Cladribine, n (%)	 65 (28.5) 23 (10) 61 (26.7) 79(34.8)
Median Charlson score of comorbidities, (IQR)	0.31 (0.41)
Median follow up time after second dose, days (IQR)	190±12
Mean follow up time after second dose, days, SD, Range	191.3±16(174-245)
Number of patients infected with SARS-CoV-2 breakthrough treated with a HET in the pre vaccinated period	42 (18.4)
First dose of SARS-CoV-2 vaccine received. Sputnik V, n (%) Astra Zeneca, n (%) Sinopharm	 65 (28.5) 115 (50.5) 48 (21)
Second dose of SARS-CoV-2 vaccine received. Sputnik V, n (%) Astra Zeneca, n (%) Sinopharm Moderna Pfizer	 47 (20.6) 126 (55.3) 34 (14.9) 11 (4.8) 10 (4.4)
Median time between first and second vaccine dose, (SD) days,	61 ±19
Homologous vaccine scheme, n (%)	194 (85)

* From first dose of the SARS-CoV-2 vaccine up to 6 months after the full course of the vaccine
RRMS= relapsing remitting multiple sclerosis; SPMS= secondary progressive multiple sclerosis; PPMS= primary progressive multiple sclerosis; EDSS= expanded disability status scale; SD= standard deviation IQR (interquartile range)

CONCLUSIONS

- We found an incidence density of breakthrough SARS_CoV-2 infection of 3.5 per 10,000 patients/day (95%CI 2.3-6.7) after vaccination in Argentina mainly with Sputnik or Astra Zeneca vaccines.
- We described the incidence rate after vaccination for every HET used, it being significantly higher for ocrelizumab compared with other HETs. We also observed a decrease in the frequency of COVID-19 infection between pre- and post-vaccination in the included HET patients.

REFERENCES: 1-Achiron, A.,et al . Humoral immune response to COVID-19 mRNA vaccine in patients with multiple sclerosis treated with high-efficacy disease-modifying therapies. Ther Adv Neurol Disord 14, 17562864211012835. 2-Golshani, M., Hrdy, J., 2022. Multiple Sclerosis Patients and Disease Modifying Therapies: Impact on Immune Responses against COVID-19 and SARS-CoV-2 Vaccination. Vaccines (Basel) 10(2). 3- Jakimovski, D.,et al., 2022. COVID-19 Vaccination in Multiple Sclerosis and Inflammatory Diseases: Effects from Disease-Modifying Therapy, Long-Term Seroprevalence and Breakthrough Infections. Vaccines (Basel) 10(5). Yamout, B.I., et al.., 2021. MENACTRIMS practice guideline for COVID-19 vaccination in patients with multiple sclerosis. Mult Scler Relat Disord 56, 103225

ACKNOWLEDGMENTS: This research was supported by Novartis Argentina. The funder of the study had no role in data collection, data analysis nor data interpretation, Novartis colaborated in the writing of the report.

DISCLOSURES: Belen Federico is an employees of Novartis. The rest of the authors have no disclosures to declare relating to this poster.

Copyright © 2023 Novartis Pharma AG. All rights reserved.

Scan this QR code to download a copy Poster

Visit the web at:http://novartis.medicalcongressposters.com/Default.aspx?doc=XXXX

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors